521-M

Phase I Study of Anti-LFA-1 Monoclonal Antibody (Cytolin®) in Adults With HIV Infection

D. Northfelt, MD[1], A. Allen[2], and P. Kapustay[2]

[1]University of California, San Diego, [2]Amerimmune, Inc.

D. Northfelt, MD
University of California, San Diego
Eisenhower Medical Center
Suite 206 Kiewit Building
39000 Bob Hope Drive
Rancho Mirage, CA92270
T: (760) 776-1633
F: (760) 776-1533
dnorthfelt@pol.net

A. Allen and P. Kapustay
Amerimmune, Inc
920 Hampshire Road, Suite A-10
Westlake Village, CA91361
T: (805) 497-7252
F: (805) 497-4732
amerimmune@earthlink.net

ABSTRACT

BACKGROUND: Human lymphocyte function-associated antigen (LFA)-1 is a heterodimeric lymphocyte surface glycoprotein. LFA-1 is expressed on CD8+ cytotoxic T lymphocytes (CTLs) prevalent in HIV-infected individuals and may be involved in the progressive depletion of CD4+ T cell counts. Anti-LFA-1 monoclonal antibodies (mAB) have been shown to reversibly inhibit CTLkilling by slowing the initial rate of cytolysis and by interfering with effector/conjugate formation. Cytolin®, a mouse anti-human mAB, is believed to bind four epitopes of the α region of LFA-1. This Phase I, single-dose study was designed to evaluate the safety and preliminary efficacy of two doses of Cytolin® in HIV-infected adults.

METHODS:Thirteen HIV-infected men (plasma HIV RNAconcentrations > 20,000 copies/mL, CD4+ T-cell counts 200-500 cells/mm3) were sequentially enrolled in two dose groups: 0.05 mg/kg (n=6) and 0.1 mg/kg (n=7). Cytolin® was administered on day 0 by slow IV infusion (0.5 mg protein/min). Hematologic, biochemical, and clinical adverse events were assessed at predetermined times after Cytolin® administration. Preliminary efficacy assessment consisted of change from baseline in HIV RNAconcentration, CD4+ and CD8+ T-cell counts. Parameters were analyzed with descriptive statistics.

RESULTS: All AEs were mild to moderate, and only one AE (diarrhea) was considered possibly related to study drug. In the 0.05 mg/kg dose group mean HIV RNA concentration was 0.265 log below baseline on Day 14 and 0.363 log above baseline on Day 56. Mean CD4+T-cell count was 37.3 cells/mm3 above baseline on Day 28 and 15 cells/mm3 below baseline on Day 56. Mean CD8+ T-cell count fluctuated, and was 153.6cell/mm3 below baseline on Day 56. In the 0.1 mg/kg dose group, mean HIV RNA concentration was 0.153 log below baseline on Day 14 and 0.229 log below baseline on Day 56. Mean CD4+ T-cell count was 20.4cells/mm3 below baseline on day 14, but 70.1 cells/mm3 above baseline on Day 56. Mean CD8+ T-cell count fluctuated, and was 107.4 cell/mm3 above baseline on Day 56.

CONCLUSIONS:Cytolin® was well tolerated. A single 0.1mg/kg dose reduced HIV RNAconcentration and increased CD4+ and CD8+ T-cell counts at 56 days. The data suggest a dose response relationship, and also suggest that higher doses currently under study may be more effective.

INTRODUCTION

Adhesion molecules are involved in different stages of HIV-1 infection and profoundly affect HIV-1 neutralization by virus-specific antibodies. In cell-to-cell interactions, the presence of LFA-1 has been shown to be crucial for virus-mediated syncytium formation. The abundant expression of adhesion molecules on CD8+ CTLs and the role of adhesion molecules in CTL-mediated lysis of target cells, suggests that LFA-1 may also be involved in the progressive depletion of CD4+ T cell counts in HIV-infected patients. In addition to the direct cytopathic effects of viral products and the direct killing by HIV-specific CTLs, other indirect mechanisms have been proposed to explain T-cell depletion. One such mechanism may be the killing of uninfected CD4+ T cells. Several studies have identified a population of non-HLA-restricted CTLs which lyse uninfected activated CD4+ lymphocytes. This population is present only in HIV seropositive individuals, and its activity has been shown to coincide with a drop in CD4+ lymphocyte numbers in vivo in some individuals.

Cytolin® is a mouse, anti-human, monoclonal antibody which binds to four epitopes of the alpha region of LFA-1. Anti-LFA-1 monoclonal antibodies have been demonstrated to reversibly inhibit CTLkilling by slowing the initial rate of cytolysis and by interfering with conjugate formation between the effector and target cells. Cytolin® is being tested for its ability to reduce CTLcytoloysis, subsequently reducing viral load in HIV-infected individuals.

Study Design

This was a single-center, open-label, single-dose evaluation of two doses of Cytolin® (0.05 and 0.1 mg/kg). Thirteen patients were sequentially enrolled in the two dose groups: 0.05 mg/kg (n=6) and 0.1 mg/kg (n=7). Just before, and at specified times after administration of Cytolin®, blood was obtained for determination of pharmacokinetic parameters, T-cells by flow cytometry, hematology, clinical chemistry, urinalysis, HIV viral load by RNAPCR,and human antimouse antibody. Karnofsky scores, physical examinations and delayed-type hypersensitivity (DTH) skin testing was also performed at pre-determined times.

Patient Population

Patients eligible for the study were male or non-pregnant females 18-years or older who were HIVpositive with a viral load greater than or equal to 20 000 copies/mLand CD4+ count of 200 to 500 cells/mm^3. Eligible patients were naive to drug therapy or had been on a fixed antiviral regimen for at least eight weeks before study participation and agreed to remain on their existing treatment throughout the study, and were able to provide written informed consent. Patients who had concurrent illness or evidence of pulmonary infection, had a history of substance abuse or other behavioral problem that prevented study participation and compliance were not eligible for the study.

DEMOGRAPHICS & BASELINE CHARACTERISTICS

Both dose groups were similar with respect to gender, ethnicity, age, and weight. No obvious differences were observed between the dose groups in baseline hematology, chemistry, or vital sign parameters.

	Dose Level		
	0.05 mg CTM/kg	0.10 mg CTM/kg	Overall
Gender, n (%)			
Male	6 (100%)	7 (100%)	13 (100%)
Female			
Ethnic Group, n (%)			
Caucasian	6 (100%)	5 (71%)	11 (85%)
Black		1 (14%)	1 (8%)
Hispanic		1 (14%)	1 (8%)
Age (yrs)			
Mean	42.7	43.6	43.2
95% CI	[37.0, 48.4]	[38.7, 48.5]	[39.7, 46.7]
SD	7.00	6.57	6.50
Median	41.6	42.4	42.4
Range	35.5, 55.2	36.4, 51.8	35.5, 55.2
Weight (kg)			
Mean	77.0	79.0	78.1
95% CI	[72.3, 81.7]	[65.7, 92.3]	[70.8, 85.4]
SD	5.97	17.9	13.3
Median	76.0	74.0	75.0
Range	70.0,85.0	53.0,110	53.0,110

SAFETY RESULTS

The overall number of adverse events was small. Thirteen subjects reported 32 adverse events. The most common adverse event was rash, which occurred in six (19%) of subjects. All adverse events were considered mild (94%) to moderate (6%). Only one adverse event (diarrhea)was considered possibly related to study drug, all other adverse events were unlikely related to study drug.

	Dose Level		
	0.05 mg CTM/kg	0.10 mg CTM/kg	Overall
Adverse Events, n	19	13	32
Adverse Events with > 20% Occurrence, n (%)			
Rash	3 (50%)	3 (43%)	6 (46%)
Insomnia	2 (33%)	0	2 (15%)
Dream Abnorm	2 (33%)	0	2 (15%)
Severity, n (%)			
Mild	17 (89%)	13 (100%)	30 (94%)
Moderate	2 (11%)	0	2 (6%)
Severe	0	0	0
Serious	0	0	0
Related to Study Drug			
Unlikely	18 (95%)	0	31 (97%)
Possibly	1 (3%)	0	1 (3%)
Probably	0	0	0
Definitely	0	0	0

EFFICACY RESULTS

Viral Load

In the 0.05 mg/kg dose group, mean HIV RNAconcentration was 2.269 log on Day 0 . HIV RNAconcentration decreased and on Day 14 was 0.265 log below baseline (2.004 log). HIV RNAconcentration increased from Day 14 to the end of study and on Day 56 was 0.363 log above baseline (2.632 log).
In the 0.1 mg/kg dose group, mean HIVRNAconcentration was 2.176 log on Day 0 and decreased 0.153 log below baseline to 2.022 log on Day 14. HIV RNA concentration increased transiently on Days 21 and 28 and was 0.229 log below baseline (1.946 log) on Day 56.

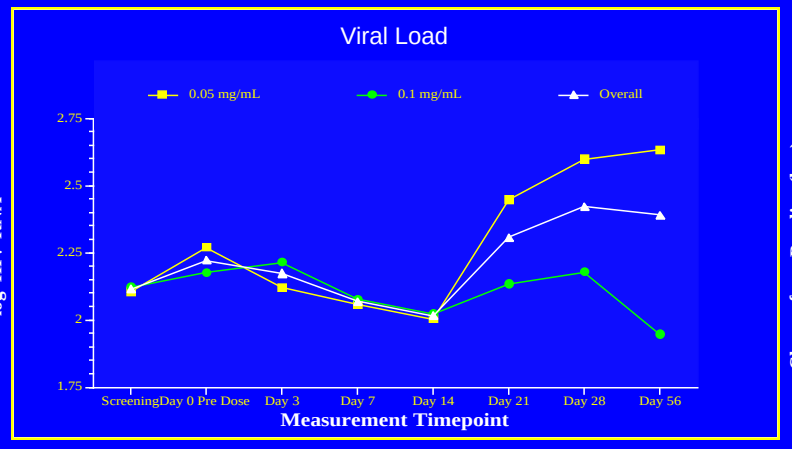

Viral Load
0.05 mg/mL
0.1 mg/mL
Overall
log HIV-RNA
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

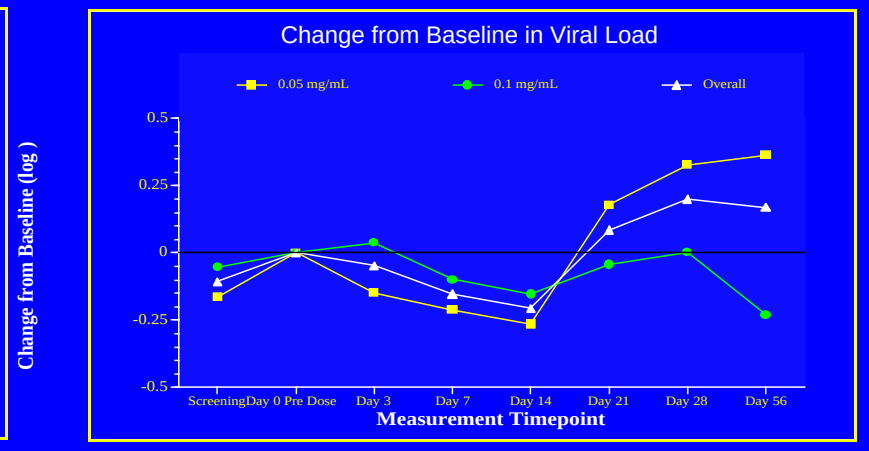

Change from Baseline in Viral Load
0.05 mg/mL
0.1 mg/mL
Overall
Change from Baseline (log)
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

CD4+ Cell Count

In the 0.05 mg/kg dose group, CD4+ T cell count at baseline was 229.3 cells/mm^3. CD4+ T-cell count decreased transiently from baseline and on Day 28 was 37.3 cells/mm^3 above baseline (266.6 cells/mm^3). At the end of the study, CD4+ T-cell count in the 0.05 mg/kg dose group was 15 cells/mm^3 below baseline (214.3 cells/mm^3).
In the 0.1 mg/kg dose group, CD4+ T cell count at baseline was 340.5 cells/mm^3. CD4+ T-cell count increased transiently from baseline and on Day 14 was 20.4 cells/mm^3 below baseline (320.1 cells/mm^3). CD4+ T-cell count increased from Day 28 to the end of study and was 70.1 cells/mm^3 above baseline on Day 56 (410.6 cells/mm^3)

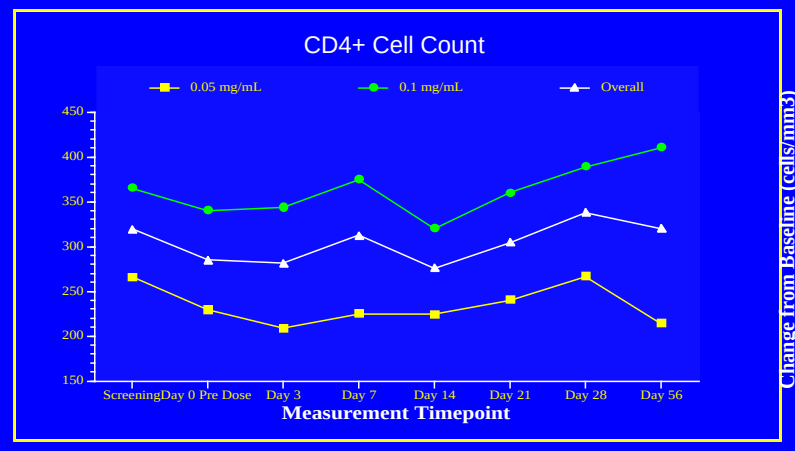

CD4+ Cell Count
0.05 mg/mL
0.1 mg/mL
Overall
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

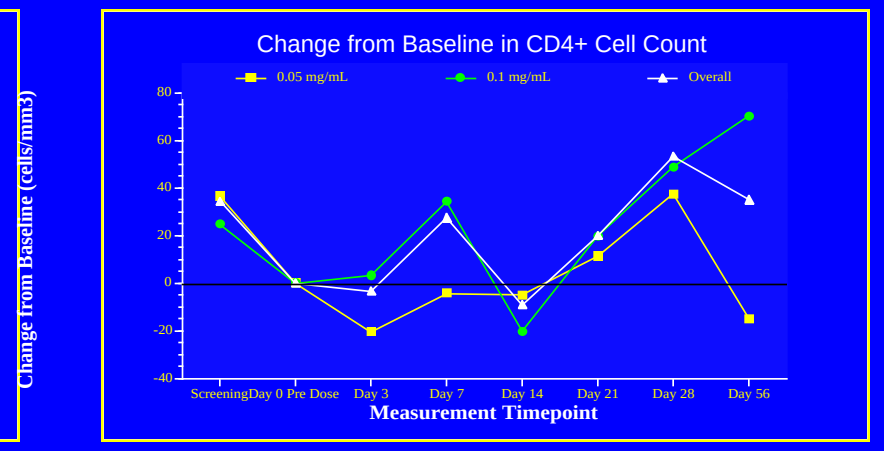

Change from Baseline in CD4+ Cell Count
0.05 mg/mL
0.1 mg/mL
Overall
Change from Baseline (cells/mm3)
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

CD8+ Cell Count

In the 0.05 mg/kg dose group, CD8+ T cell count at baseline was 1205.3 cells/mm^3. CD8+ T-cell count fluctuated and at the end of study (Day 56) was 153.6 cells/mm^3 below baseline (1051.7 cells/mm^3).
In the 0.1 mg/kg dose group, CD8+ T cell count at baseline was 1117.2 cells/mm^3. CD4+ T-cell count increased transiently from baseline and on Day 14 was 20.8 cells/mm^3 below baseline (1096.4 cells/mm^3). CD4+ T-cell count increased from Day 28 to the end of study and was 107.4 cells/mm^3 above baseline on Day 56 (1224.6 cells/mm^3).

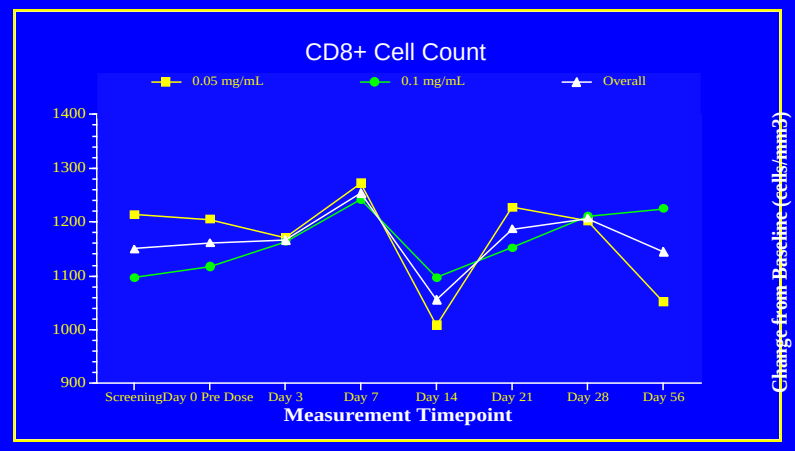

CD8+ Cell Count
0.05 mg/mL
0.1 mg/mL
Overall
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

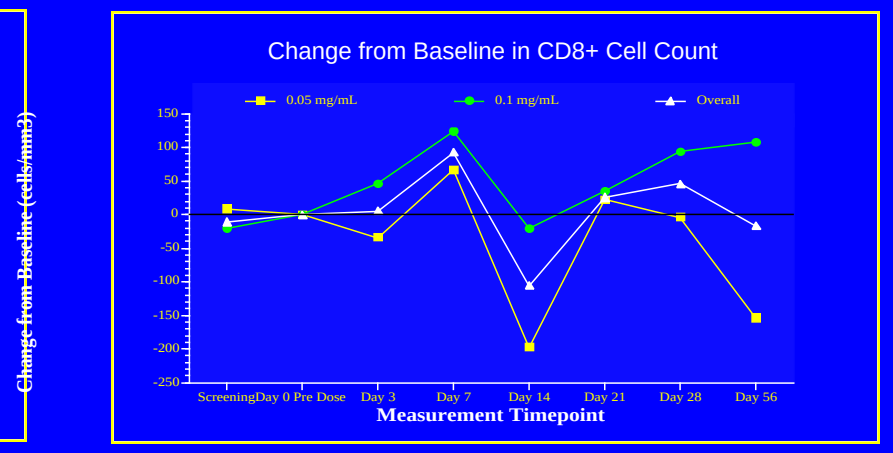

Change from Baseline in CD8+ Cell Count
0.05 mg/mL
0.1 mg/mL
Overall
Screening Day 0 Pre Dose Day 3 Day 7 Day 14 Day 21 Day 28 Day 56
Measurement Timepoint

Conclusions

Cytolin was safe and well tolerated in this study. All adverse events were mild to moderate in severity and only 1 adverse event (diarrhea)was possibly related to study medication. No subjects reported serious adverse events and no subjects withdrew from the study. Efficacy results suggest a dose response relationship of Cytolin®. At the end of study (Day 56) in the 0.1 mg/kg dose group, viral load decreased from baseline, and CD4+ and CD8+ T-cell count increased from baseline suggesting that Cytolin® may be reducing lysis of CD4+ T-cells thus reducing HIV viral load. Ongoing studies investigating multiple dosing with higher doses may demonstrate a linear relationship between dose and efficacy and suggest that multiple dosing at these higher doses may be more effective in boosting the immune system and reducing HIV viral load.