GDS-044 Van Doren 2 2/22/02 5:23 PM Page 1

Poster Number

415-W

9th Conference on Retroviruses and Opportunistic Infections
February 24-28, 2002
Seattle, Washington

The Viread™ expanded access program: safety and efficacy of tenofovir disoproxil fumarate in antiretroviral treatment-experienced patients

S. Follansbee[1], J. Reynes[2], M. Nelson[3], B. Clotet[4], A. Lazzarin[5], A. Adam[6], S. Van Doren[7], R. Buffels[7], S. Barriere[7], L. Zagury[7], I. Miranski[7], G. Choy[7], J. Rooney[7]

[1]Kaiser Permanente Medical Center, San Francisco, California, USA, [2]Hôpital Gui de Chauliac, Montpellier, France, [3]Chelsea and Westminster Hospital, London, UK
[4]Hospital Germans Trias I Pujol, Barcelona, Spain, [5]Ospedale San Raffaele, Milano, Italy, [6]IPM-Study Center, Hamburg, Germany, [7]Gilead Sciences, Foster City, California, USA, and Paris, France

Stephen Follansbee, MD
Kaiser Permanente
Clinical Trials Unit
4141 Geary Blvd, Suite 219
San Francisco, CA 94118 USA
Office (415) 833-3480
Fax (415) 833-3483
Stephen.Follansbee@KP.org

Introduction

- Viread™ (tenofovir disoproxil fumarate [DF]) is a once-daily nucleotide reverse transcriptase inhibitor (NtRTI) indicated in combination with other antiretroviral agents for the treatment of HIV-1 infection. Administered once daily as a single 300-mg tablet, Viread™ has additive or synergistic activity with other antiretrovirals in vitro and has activity in resting and activated T cells. In 2 controlled clinical trials conducted in treatment-experienced adults with evidence of HIV-1 viral replication despite ongoing antiretroviral therapy, Viread™ added to background therapy provided sustained viral suppression through 48 weeks. The most common adverse effects that occurred in patients receiving Viread™ with other antiretroviral agents in clinical trials were mild to moderate gastrointestinal events, such as diarrhea, vomiting, and nausea. Less than 1% of patients discontinued participation in the clinical studies due to gastrointestinal adverse events. Laboratory abnormalities observed in these studies occurred with similar frequency in the Viread™ and placebo-treated groups[1]

- Prior to commercial availability, a global expanded access program (EAP) was initiated in March 2001 for HIV-1 infected patients who failed prior highly active antiretroviral therapy (HAART) and had limited treatment options. To be eligible to participate, patients must have met the following criteria prior to dosing: age ≥18 years; confirmed laboratory diagnosis of HIV-1 infection; unable to construct a viable treatment regimen without Viread™; had no hematologic, renal, or hepatic dysfunction; was not pregnant or breast-feeding; and agreed to use an effective barrier contraceptive method while receiving Viread™. Initial study entry criteria included plasma HIV-1 RNA ≥10,000 copies/mL by polymerase chain reaction assay and CD4 counts ≤100 cells/mm³ or CD4 count >100 and ≤200 cells/mm³ with a history of an AIDS-defining opportunistic infection, and failure of at least 2 protease inhibitors (PIs) or at least 1 PI plus a non-nucleoside reverse transcriptase inhibitor (NNRTI); however, these specific criteria were removed shortly after the start of enrollment in countries that specified these entry criteria. As of January 14, 2002, 7317 patients were enrolled in the EAP globally: 3453 (United States), 1635 (France), 722 (United Kingdom), 790 (Spain), 344 (Germany), 176 (Australia), 73 (Netherlands), 46 (Canada), 41 (Italy), 37 (Portugal, Ireland). Data are available on all patients for serious adverse events (SAEs). Baseline demographics on 2750 patients in the United States (January 13, 2002) and 2289 patients in Europe/Australia (December 15, 2001) are presented. Efficacy data for France, Italy, and Germany are presented. EAPs were closed following marketing approval for Viread™ in each country (November 2001 for the US program and February 2002 for France, UK, and Germany)

Objective

- Evaluate the safety and efficacy of Viread™ in antiretroviral-experienced patients enrolled in the Viread™ Global EAP

Methods

Design

- Open-label protocol or treatment guidelines according to local regulations in each country (see Table 1)

- Viread™ 300 mg orally once daily with other antiretroviral agents in patients failing HAART. Physicians were encouraged to add at least 1 active antiretroviral agent in addition to Viread™ in constructing a new regimen

Table 1 — Expanded access programs

Country	Program Type	Start Date	Stop Date
United States	Protocol	March 2001	November 2001
France	Autorisation Temporaire d' Utilisation (ATU)	March 2001	February 2002
United Kingdom	Treatment Guidelines	March 2001	February 2002
Spain	Treatment Guidelines	May 2001	Ongoing
Germany	Protocol	July 2001	February 2002
Italy	Protocol	July 2001	Ongoing
Ireland	Treatment Guidelines	August 2001	Ongoing
Portugal	Treatment Guidelines	September 2001	Ongoing
Netherlands	Treatment Guidelines	October 2001	Ongoing
Australia	Special Access Scheme (SAS)	October 2001	Ongoing
Canada	Protocol	October 2001	Ongoing

Figure 1 — Total enrollment by country

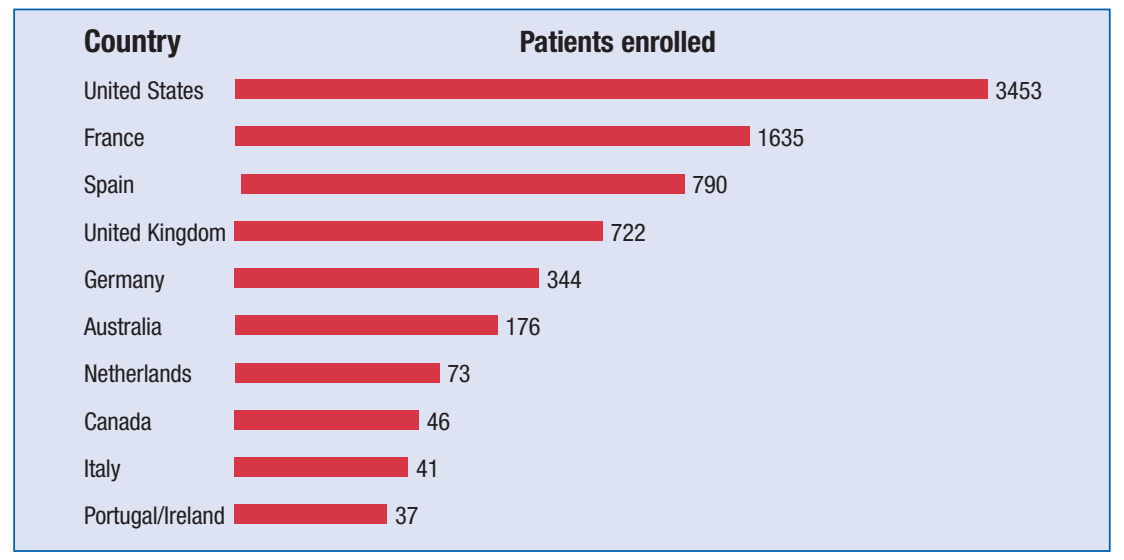
Country
Patients enrolled
United States
3453
France
1635
Spain
790
United Kingdom
722
Germany
344
Australia
176
Netherlands
73
Canada
46
Italy
41
Portugal/Ireland
37

Schedule of assessments

- Varied by country according to local regulations but was generally at baseline, month 1, month 3, and every 6 months (Europe [EU]), or at baseline, month 1, and every 2 months (North America [NA–US/Canada])

Safety assessments (NA, EU, and Australia)

- SAEs—all programs
 - — A serious adverse event is defined as an event that results in death, a life-threatening situation, inpatient hospitalization or prolongation of hospitalization, persistent or significant disability, or congenital anomaly
- Adverse events (AEs)—France (related AEs only), Germany, and Italy
- Laboratory abnormalities—France (selected labs), Germany, and Italy

Efficacy assessments (EU and Australia)

- Change in CD4 count (cells/mm3) and HIV-RNA from baseline (log_{10} copies/mL)—France, Germany, and Italy

Key inclusion criteria

- Documented diagnosis of HIV-1 infection
- Treating physician is unable to construct a viable combination of antiretroviral agents
- Negative pregnancy tests
- Willingness to use effective contraception

Key exclusion criteria

- Patients receiving ongoing nephrotoxic agents or within 7 days of study enrollment
- Pregnant women or women who are breast-feeding
- Current alcohol or substance abuse
- History of clinically significant renal or bone disease

Results

As of January 14, 2002, a total of 7317 subjects were enrolled in the Viread™ Global EAP. Enrollment began in March 2001 in US, France, and the UK and was further extended to other countries as noted in Table 1. Programs were closed in the US in November 2001 and in France, UK, and Germany in February 2002 following approval for marketing in those countries. Programs continued in other countries. Baseline demographic data are available on 2750 patients in the US and 2289 patients in Europe/Australia

Table 2 — Summary of demographic and baseline disease characteristics

Baseline Demographics	US	EU/Australia	France	UK	Spain	Germany
Patients with baseline visit	2750	2289	785	606	419	231
Median age (years)	42	40	40	39	38	42
Gender						
Male	90%	79%	80%	72%	79%	87%
Female	10%	18%	20%	17%	21%	13%
Missing data	0%	3%	0%	11%	0%	0%
Median weight (kg)	74	67	65	69	66	72
CD4 count (cells/mm³)						
<50	11%	16%	21%	14%	11%	9%
50–200	11%	29%	30%	33%	21%	20%
>200	9%	52%	49%	41%	68%	71%
Not specified	70%	3%	0 %	12%	0%	0%
Median	90	222	194	190	334	254
HIV RNA (copies/mL)						
≤500	1%	19%	8%	18%	37%	35%
>500–10,000	3%	17%	16%	17%	17%	22%
>10,000–100,000	9%	32%	40%	25%	26%	27%
>100,000	12%	26%	35%	20%	19%	16%
Not specified	75%	6%	1%	20%	1%	0%
Median (log_{10} copies/mL)	4.98	4.42	4.75	4.22	3.70	3.63
Median time from HIV diagnosis (months)	NR	117	126	95	111	102

NR=not reported; EU=all countries participating: France, Spain, UK, Germany, Netherlands, Italy, Portugal, and Ireland

Table 3 — Duration of treatment with Viread™

Time on Viread™ (weeks)	US	France	Spain	UK	Germany
Mean	10.2	22.0	18.9	22.0	11.6
Median	9.0	22.7	20.1	23.1	10.7
Range	1.0–36.0	0.6–40.7	1.4–28.7	0.7–40.6	0.9–20.7

Table 4 — Discontinuation of Viread™

All Countries (n=5039)	
Total number of discontinuations	**315 (6%)**
Adverse event	101 (2%)
Death	29 (0.6%)
Disease progression	24 (0.5%)
Patient request removal	23 (0.5%)
Patient noncompliance	20 (0.4%)
Lost to follow-up	55 (1%)
Other	63 (1%)

Table 5 — Summary of antiretroviral (ART) medication use

Number of ART	US		EU/Australia		France		Spain		Germany	
N	2750		2289		785		419		231	
	Previous	Baseline	Previous	Baseline	Previous	Baseline	Previous	Baseline	Previous	Baseline
Missing	--*	19%	27%	24%	0%	1%	0%	7%	0%	0%
1	--	7%	0%	--	0%	--	0%	--	0%	--
2	--	21%	0%	1%	0%	1%	0%	1%	0%	3%
3	--	29%	2%	30%	1%	46%	4%	35%	2%	31%
4	--	17%	2%	27%	3%	34%	2%	24%	4%	39%
5	--	5%	3%	14%	3%	12%	4%	24%	4%	20%
6	--	2%	6%	4%	6%	4%	12%	7%	8%	6%
7	--	<1%	7%	1%	10%	2%	9%	2%	9%	1%
8	--	--	9%	--	12%	--	13%	--	12%	--
9	--	--	9%	--	12%	--	10%	--	9%	--
10	--	--	9%	--	13%	--	12%	--	9%	--
11	--	--	8%	--	13%	--	10%	--	11%	--
12	--	--	8%	--	11%	--	11%	--	13%	--
13	--	--	6%	--	8%	--	6%	--	7%	--
14	--	--	4%	--	5%	--	4%	--	11%	--
15	--	--	1%	--	1%	--	1%	--	2%	--

* Data not collected

Table 6 — Most common prior and baseline ARTs

Antiretroviral Medications	US		EU/Australia		France		Spain		Germany	
	Prior	Baseline	Prior	Baseline	Prior	Baseline	Prior	Baseline	Prior	Baseline
N	2286	2286	1680	2289	785	785	419	419	231	231
Protease Inhibitors										
Indinavir	52%	7%	72%	3%	75%	4%	74%	2%	62%	2%
Nelfinavir	50%	5.0%	60%	2%	64%	2%	60%	3%	52%	3%
Ritonavir	47%	15%	67%	11%	69%	15%	64%	19%	63%	12%
Saquinavir	48%	7%	62%	4%	61%	5%	60%	6%	60%	7%
Amprenavir	28%	24%	24%	16%	31%	15%	5%	37%	30%	15%
Lopinavir/ritonavir	13%	53%	38%	45%	35%	71%	32%	42%	57%	54%
Nucleoside Reverse Transcriptase										
Stavudine	50%	32%	90%	11%	90%	6%	91%	22%	89%	14%
Zidovudine	81%	12%	91%	9%	93%	11%	87%	6%	91%	30%
Abacavir	35%	26%	61%	24%	60%	30%	55%	29%	76%	42%
Didanosine	43%	28%	87%	28%	91%	26%	86%	56%	83%	32%
Zalcitabine	27%	2%	49%	5%	56%	12%	43%	1%	46%	2%
Lamivudine	69%	42%	95%	36%	95%	51%	93%	37%	96%	59%
Non-nucleoside Reverse Transcriptase										
Delavirdine	13%	4%	4%	1%	5%	1%	<1%	<1%	8%	3%
Nevirapine	39%	8%	61%	8%	56%	6%	62%	22%	62%	10%
Efavirenz	41%	17%	55%	13%	56%	18%	52%	18%	68%	15%
Nucleotide Analog										
Tenofovir disporoxil fumarate	0%	2%	0%	0%	0%	0%	0%	0%	2%	0%
Adefovir dipivoxil	8%	<1%	0%	0%	2%	0%	0%	0%	0%	0%
Other										
Hydroxyurea	21%	3%	16%	1%	13%	1%	24%	1%	8%	<1%
T20	<1%	3%	7%	3%	<1%	4%	1%	5%	2%	1%

Figure 2 — Mean change in HIV-1 RNA (log_{10} copies/mL) from baseline

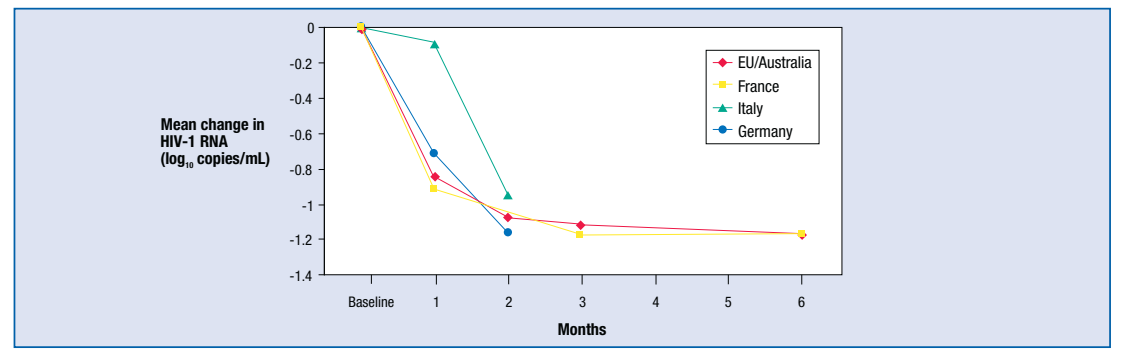
Mean change in HIV-1 RNA (log_{10} copies/mL)
0
-0.2
-0.4
-0.6
-0.8
-1
-1.2
-1.4
Baseline
1
2
3
4
5
6
Months
EU/Australia
France
Italy
Germany

Figure 3 — Mean change in CD4 (cells/mm³) count from baseline

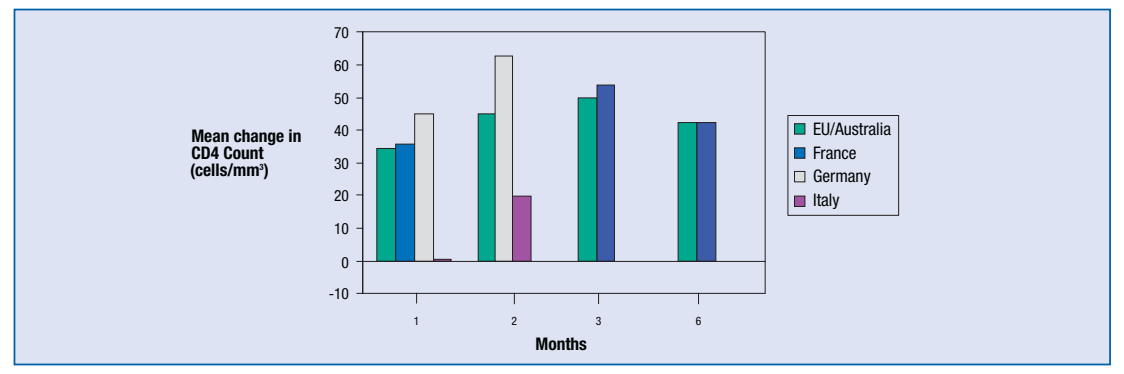
Mean change in CD4 Count (cells/mm³)
70
60
50
40
30
20
10
0
-10
1
2
3
6
Months
EU/Australia
France
Germany
Italy

Table 7 — Summary of serious adverse events by body system classification

	United States N=3453		EU/Australia N=3818	
Body system	All SAEs	Related SAEs	All SAEs	Related SAEs
Body as a whole	105 (4%)	9 (<0.3%)	32 (1%)	6 (<0.2%)
Cardiovascular system	32 (1%)	5 (<0.1%)	4 (<0.1%)	1 (<0.1%)
Digestive system	91 (3%)	21(<1%)	20 (1%)	11(<0.3%)
Hemic and lymphatic system	33 (1%)	2 (<0.1%)	13 (<0.3%)	9 (<0.2%)
Metabolic and nutritional disorders	36 (1%)	11(<1%)	7 (<0.2%)	6 (<0.2%)
Musculoskeletal system	9 (<0.3%)	0 (0%)	4 (<0.1%)	3 (<0.1%)
Nervous system	32 (1%)	5 (<0.1%)	12 (<0.3%)	6 (<0.2%)
Respiratory system	41 (1%)	1(<0.1%)	7 (<0.2%)	1 (<0.1%)
Skin and appendages	6 (<0.2%)	1 (<0.1%)	6 (<0.2%)	3 (<0.1%)
Special senses	4 (<0.1%)	0 (0%)	2 (<0.1%)	1 (<0.1%)
Urogenital system	28 (1%)	10 (<0.3%)	12 (<0.3%)	8 (<0.2%)
Total	417 (12%)	65 (2%)	119 (3%)	55 (1%)

- Bone disease or fractures were reported in 0.1% of patients. Adverse events were reported infrequently in programs where AEs were collected. In the French EAP, related adverse events were reported in 74 patients (5%) with GI events being most common (2%)
- Serum creatinine and phosphorus were routinely collected at each study visit in the French and German Programs (see Table 8)

Table 8 — Maximum grade toxicity of serum creatinine and phosphorus

Serum Creatinine (mg/dL)	France	Germany
Missing	0 (0%)	0 (0%)
No abnormality	765 (98%)	231 (100%)
1 ≥0.5 over baseline	18 (2%)	0 (0%)
2 2.1–3.0	1 (0.1%)	0 (0%)
3 3.1–6.0	1 (0.1%)	0 (0%)
4 >6.0	0 (0%)	0 (0%)
Hypophosphatemia[1] (mg/dL)		
Missing	0 (0%)	2 (1%)
No abnormality	638 (81%)	207 (90%)
1 2.0–2.2	104 (13%)	18 (8%)
2 1.5–1.9	39 (5%)	2 (1%)
3 1.0–1.4	4 (1%)	2 (1%)
4 <1.0	0 (0%)	0 (0%)

Conclusions

- As of January 14, 2002, a total of 7317 patients have been enrolled in 11 countries in the Viread™ Global EAP
- Participants in the Viread™ Global EAP were heavily pretreated with NRTIs, NNRTIs, and PIs with the majority having HIV-1 RNA ≥10,000 copies/mL at baseline
- The safety and tolerability profile of Viread™ in the EAP was similar to that observed in controlled clinical trials. Serious related adverse events were reported in 2% of patients in the US EAP and 1% in the EU/Australia EAP with all individual SAEs occurring in <1% of patients
- Abnormalities in renal function were infrequent and were similar to the rates reported in controlled clinical trials in Viread™ and placebo patients
- Reductions in HIV RNA and increases in CD4 counts from baseline were observed through 6 months in these heavily ART-experienced patients treated with Viread™ in combination with other antiretroviral agents

Acknowledgements

- Gilead Sciences, Inc would like to thank the large number of patients, physicians, and their research personnel who have participated in of the Viread™ Global EAP

References

Viread™ (tenofovir disoproxil fumarate) [package insert]. Foster City Calif: Gilead Sciences, Inc; October 26, 2001