GDS-043 Miller 48wk genotypic 2/20/02 11:13 AM Page 1

Poster Number
414-W
9th Conference on Retroviruses and Opportunistic Infections
February 24-28, 2002
Seattle, Washington

Final 48-week genotypic and phenotypic analyses of Study 907: tenofovir DF added to stable background regimens

N.A. Margot, A. Johnson, D.F. Coakley, A.K. Cheng, M.D. Miller
Gilead Sciences, Foster City, California, USA

Michael Miller
Gilead Sciences
333 Lakeside Drive
Foster City, CA 94404
(650) 522-5584
Fax (650) 522-5890
michael_miller@gilead.com

Introduction

Tenofovir disoproxil fumarate (Viread™)

Tenofovir DF has the following characteristics:

- Unique acyclic nucleotide analog with phosphonate group
- Anti-HIV activity in resting and activated T cells and macrophages
- Weak inhibitor of mitochondrial polymerase γ (K_i >50 µmol/L, selectivity index >3000)

In vitro resistance profile of tenofovir

- Tenofovir can select in vitro for the K65R mutation in RT
 - 3- to 4-fold reduced susceptibility to tenofovir
 - K65R also selected by ddI, ddC, and abacavir
 - Low prevalence in antiretroviral-experienced patients (<2%)
- Tenofovir retains activity against most nucleoside-resistant forms of HIV-1
- Increased activity against HIV-1 expressing the M184V mutation

Objective

- Characterize and evaluate tenofovir DF resistance at week 48 in Study 907

Methods

Study design

- Randomized, double-blind, placebo-controlled intensification study
- Entry criteria
 - HIV RNA 400 to 10,000 copies/mL
 - Stable ART ≥8 weeks prior to entry consisting of ≤4 concomitant antiretroviral agents
 - No CD4 cell count restrictions

Figure 1 — Study 907 schema chart

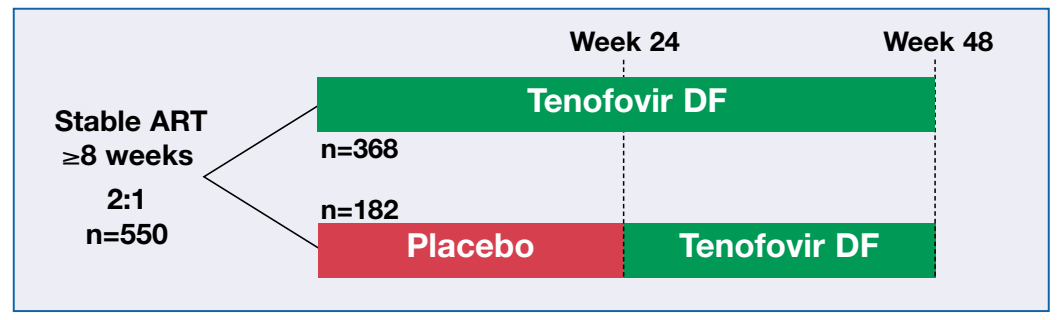
Week 24
Week 48
Stable ART
≥8 weeks
2:1
n=550
Tenofovir DF
n=368
n=182
Placebo
Tenofovir DF

Patient baseline characteristics

- Median HIV-1 RNA: 2340 copies/mL
- Mean CD4 count: 426 cells/mm³
- Mean prior HIV treatment: 5.4 years

Resistance analyses

- 50% of patients randomly assigned to genotyping substudy (n=274)
 - Genotypic analyses (Virco) at baseline, week 24 and week 48, or early termination
 - Baseline data available for 253 patients
 - Week 48/early termination data available for 145 patients
 - Remaining patients have insufficient HIV RNA for analysis
- 25% of patients randomly assigned to phenotyping substudy (n=137)
 - Phenotypic analyses (Virco) at baseline and week 48, or early termination
 - Baseline data available for 109 patients
 - Matching week 48 data available for 59 patients
 - Remaining patients have insufficient HIV RNA or virus for analysis

Results

Table 1 — Development of nucleoside-associated HIV mutations by week 48 (genotyping substudy, n=253)

RT Resistance Mutations Developing	Concomitant Nucleoside ART	Percentage of Patients: Placebo-controlled Phase (Weeks 0-24) Placebo (n=84)	Placebo-controlled Phase (Weeks 0-24) Tenofovir DF (n=169)	Open-label Phase (Weeks 24-48) Tenofovir DF (n=253)
Any nucleoside-associated mutation (NAM)		24% (20)	16% (27)	23% (58)
Any TAM*		14% (12)	11% (19)	19% (48)
M41L	d4T, ZDV, ddI, ABC, 3TC	4% (3)	5% (8)	5% (13)
K70R/Q/N	d4T, ZDV, ddI, 3TC	4% (3)	4% (6)	2% (6)
D67N	d4T, ZDV, ddI, 3TC	1% (1)	4% (7)	6% (16)
T215Y/F/I	d4T, ZDV, ddI, ABC, 3TC	6% (5)	2% (3)	4% (11)
L74V/I	d4T, ddI, ABC, 3TC	6% (5)	<1% (1)	3% (7)
K65R	d4T, ZDV, ddI, ABC, 3TC	—	3% (5)	1% (3)
K219E/Q/R	d4T, ZDV, ddI, ABC, 3TC	2% (2)	1% (2)	6% (15)
L210W/S	d4T, ddI	1% (1)	1% (2)	3% (8)
M184V	ZDV, ABC, 3TC	2% (2)	—	<1% (2)
T69N/I	d4T, ddI	1% (1)	<1% (1)	<1% (2)
V75I/A	d4T, ddI, ABC	1% (1)	<1% (1)	<1% (1)
A62V	ZDV, 3TC	1% (1)	—	<1% (1)
Y115F	d4T, ABC, 3TC	—	<1% (1)	<1% (2)
Q151M	d4T, ABC, 3TC	—	<1% (1)	—
T69 Insertion	—	—	—	—

* Thymidine analog-associated mutations at RT codons 41, 67, 70, 210, 215, or 219.

Table 2 — HIV RNA responses in patients developing mutations during the open-label phase (ITT)

Mutation Developing*	N	Mean DAVG48†‡	Mean Absolute Change from Baseline to Week 48‡
D67N/G	16	-0.56	-0.28
K219Q/R/E/N	15	-0.60	-0.35
M41L	13	-0.99	-0.56
T215F/Y/I/S	11	-0.83	-0.38
L210W	8	-0.31	-0.22
L74V/I	7	-0.09	0.29
K70R	6	-0.87	-0.55

* Mutations developing in >2% of patients.
† Average change in HIV RNA from baseline to week 48 ($\log_{10}$ copies/mL).
‡ Change from week 24 to week 48 was used for placebo patients ($\log_{10}$ copies/mL).

Table 3 — HIV RNA responses and tenofovir susceptibility in patients developing K65R mutation by week 48 (n=8)

Patient Code	Time of Detection	Mean DAVG48*†	Mean Absolute Change from Baseline to Week 48†	Tenofovir Susceptibility at Week 48‡
A	Week 48	0.53	1.65	0.4
B	Week 24	-0.01	-0.01	1.2
C	Week 48	-0.46	0.00	NA
D	Week 48	-1.15	-0.68	NA
E	Week 24	0.03	0.08	1.0
F	Week 24	-1.01	-1.27	NA
G	Week 24	-1.01	-1.32	0.4
H	Week 24	0.86	1.20	6.4
	Averages:	-0.28	-0.04	1.9

* Average change in HIV RNA for baseline to week 48 ($\log_{10}$ copies/mL).
† Change from week 24 to week 48 was used for placebo patients ($\log_{10}$ copies/mL).
‡ Fold change compared to wild-type control.
NA=not available, insufficient PCR product or virus for analysis.

Table 4 — Week 48 phenotypic analysis (n=59)

Patients Developing New NRTI Mutation	N	Mean Fold Change in Susceptibility from Baseline: Tenofovir	ZDV	d4T	ddI	3TC	ABC
All tenofovir DF patients*	59	1.3	2.1	1.6	2.3	1.4	1.6
No mutations by week 48	29	1.0	1.2	1.3	1.4	0.9	0.9
Yes, mutations by week 48	30	1.7	3.0	1.8	3.3	1.9	2.2
K65R developer	5	1.7	1.2	1.0	1.3	2.8	1.1

* 12 placebo-treated patients rolled over to tenofovir DF at week 24 are included. Week 48 phenotype compared with week 24 (n=9) or baseline (n=3).

- 4 of 59 patients had a greater than 3-fold change in tenofovir DF susceptibility at week 48
 - 3 patients remained within the normal tenofovir DF range (<3-fold of wild-type) due to baseline tenofovir DF hypersusceptibility
 - 1 patient had reduced susceptibility (6.4-fold) in association with K65R mutation

Table 5 — Genotypic and phenotypic changes in tenofovir DF patients showing HIV RNA rebound*

Patient	Resistance Mutations Developing: In RT	In Protease	Tenofovir Susceptibility at Week 48†
#1	M41L, T215Y	None	NA
#2	NA	NA	NA
#3	G190A	None	NA
#4	M41L, A62V/A, T215S/F	V32I/V	0.3
#5	None	None	0.4
#6	M41L, T215F	None	1.3
#7	K219R/K	L90M	0.2
#8	NA	NA	NA

* 8/41 tenofovir DF-treated patients whose HIV RNA was <50 copies/mL at week 24 had >500 copies/mL at week 48 and were classified as rebounders.
† Expressed in fold change from wild-type control.
NA=not available, insufficient PCR product or virus for analysis.

Conclusions

- At 48 weeks, development of TAMs and NAMs during the open-label phase was similar to the placebo-controlled phase, suggesting background ART is responsible for their development
 - Continued HIV RNA suppression in most patients developing new mutations
- Low incidence of mutations associated with tenofovir DF therapy through 48 weeks (3% with K65R, 8 of 253 patients)
 - Highly variable response in patients developing K65R, with 3 of 8 patients maintaining greater than 0.68 log HIV RNA suppression
 - Low-level (<2-fold) tenofovir DF phenotypic resistance associated with K65R in 4 of 5 evaluable patients
 - Full phenotypic susceptibility to d4T or AZT in patients developing K65R
 - No TAMs present in any patient developing K65R
- 1 of 59 analyzed patients developed greater than 3-fold phenotypic resistance to tenofovir DF at week 48
 - Associated with K65R
- HIV RNA rebound not associated with development of resistance to tenofovir DF
 - Resistance to other agents observed in most cases
- Phenotypic or genotypic resistance to tenofovir DF is infrequent after up to 48 weeks of tenofovir DF exposure